CVI-LM001, a first-in-class novel small molecule PCSK9 modulator for hypercholesterolemia and NASH: preclinical and first-in-human studies

Jingwen Liu[1], Bo Jiang[2], Shuiping Zhao[3], Siyu Cai[2], Minghui Huang[1] , Pingfei Fang[3], Zourong Ruan[2], Minli Chen[4], Qiyang Sho[4] , Francois Briand[5], Jianan Wang[2]

[1] CVI Pharmaceuticals Limited, Shanghai, China; [2] The Second Affiliated Hospital of Zhejiang University School of Medicine, Hangzhou, China; [3] The Second Xiangya Hospital of Central South University, Changsha, China; [4] Animal Experimental Research Center, Zhejiang University of Traditional Chinese Medicine, Hangzhou, China; [5] Physiogenex, Escalquens, France

PREMISE

- NAFLD/NASH and cardiovascular disease are associated with common metabolic risk factors such as hyperlipidemia.
- CVI-LM001, currently at Phase 2 Proof-of-Concept clinical development stage for the treatment of hypercholesterolemia, is a novel first-in-class oral small molecule PCSK9 modulator discovered and developed by CVI Pharmaceuticals.
- In preclinical studies, CVI-LM001 treatment of hamsters fed a high fat and high cholesterol diet (HFHCD) increased protein levels of LDL receptor and activated AMPK in liver tissues and decreased circulating PCSK9 levels concomitant with reductions of serum LDL-C, TC and TG levels.
- The studies aimed to demonstrate the effects of CVI-LM001 on hepatic steatosis and NASH in hamster models and conduct first-in-human Phase I clinical trials to demonstrate the safety and pharmacological activity of CVI-LM001 in Chinese healthy subjects and hyperlipidemic subjects.

METHODS

- The effects of CVI-LM001 on hepatic steatosis were evaluated in high fat and high cholesterol diet (HFHCD)-fed hamsters following 4 weeks of treatment by oral gavage at CVI-LM001 dose of 10 mg/kg, 20 mg/kg or 40 mg/kg, or 50 mg/kg fenofibrate, or vehicle once daily (QD).
- The effects of CVI-LM001 on hepatic inflammation, ballooning and fibrosis were evaluated in a diet-induced NASH hamster model following 5 weeks of treatment. Hamsters were fed a free choice diet (free choice between a chow diet with normal tap water or a high fat/cholesterol diet (Safe Diets) with 10% fructose enriched tap water) for up to 20 weeks (15 weeks diet and 5 weeks treatments of vehicle, CVI-LM001 at 100 mg/kg, or elafibranor 15 mg/kg orally QD).
- Double-blind, randomized single ascending-dose (100-800 mg per day) and multiple ascending-dose (MAD) studies were conducted to assess the tolerability, pharmacokinetics and pharmacodynamics of oral CVI-LM001 in healthy subjects. Subjects (Chinese males or females , 18-45 years) received 100, 200, or 300 mg once daily CVI-LM001 or placebo for 10 days. Blood for pharmacokinetics and/or pharmacodynamics were collected. Serum PCSK9 levels were measured.
- To explore the efficacies of CVI-LM001 in LDL-C lowering and serum PCSK9 reduction, a randomized, double-blind and placebo-controlled proof of mechanism (POM) study was further conducted in Chinese subjects with mild hypercholesterolemia (LDL-C ≥ 3.2 mM and ≤4.88 mM). Hyperlipidemic subjects (males or females , 18-65 years) received 200 mg (n=10), or 300 mg (n=11) once daily CVI-LM001 or placebo (n=11) for 28 days. Blood for pharmacokinetics and/or pharmacodynamics were collected. Serum PCSK9 and lipid levels including total cholesterol (TC), LDL-C, and ApoB were measured.

RESULTS SUMARY

- CVI-LM001 demonstrated dose-dependent reductions in serum LDL-C, TC and TG of hyperlipidemic hamsters with an effective dose of 20 mg/kg that lowered LDL-C by 37%, TC by 39% and TG by 40% compared to vehicle and ameliorated the HFHCD-induced liver fat accumulation evidenced by a 67% (p<0.01) reduction of Oil red-O staining intensity compared to vehicle.
- In the NASH model, CVI-LM001 treatment improved total NASH score (-2.1, p<0.01) driven primarily by substantial reductions of hepatic ballooning (-73%, p<0.001).
- CVI-LM001 was safe and well tolerated in healthy human volunteers at all doses evaluated. Circulating PCSK9 levels were reduced in healthy volunteers after 10 days of treatment.
- Moreover, in hyperlipidemic subjects, compared to placebo, LDL-C and PCSK9 levels were significantly reduced by CVI-LM001 at a daily dose of 300 mg after 28 days of treatment.

Preclinical Study Results

Figure 1. Significant reductions of serum TC, LDL-C, TG and liver fat accumulations by oral treatment of HFHCD-fed golden Syrian hamsters with 20 mg/kg and 40 mg/kg CVI-LM001 for 4 weeks (n=10 per group, fenofibrate (FENO) at 50 mg/kg was used as a positive control **(A).** In **B**, frozen liver sections were stained with Oil red-O and the intensity of each section was quantified (n=10 livers per group). In a parallel PK study of hamsters treated with CVI-LM001 at 40 mg/kg, CVI-LM001 showed similar plasma concentrations at day 1 and day 28, indicating no drug accumulation in this hamster model **(C).**

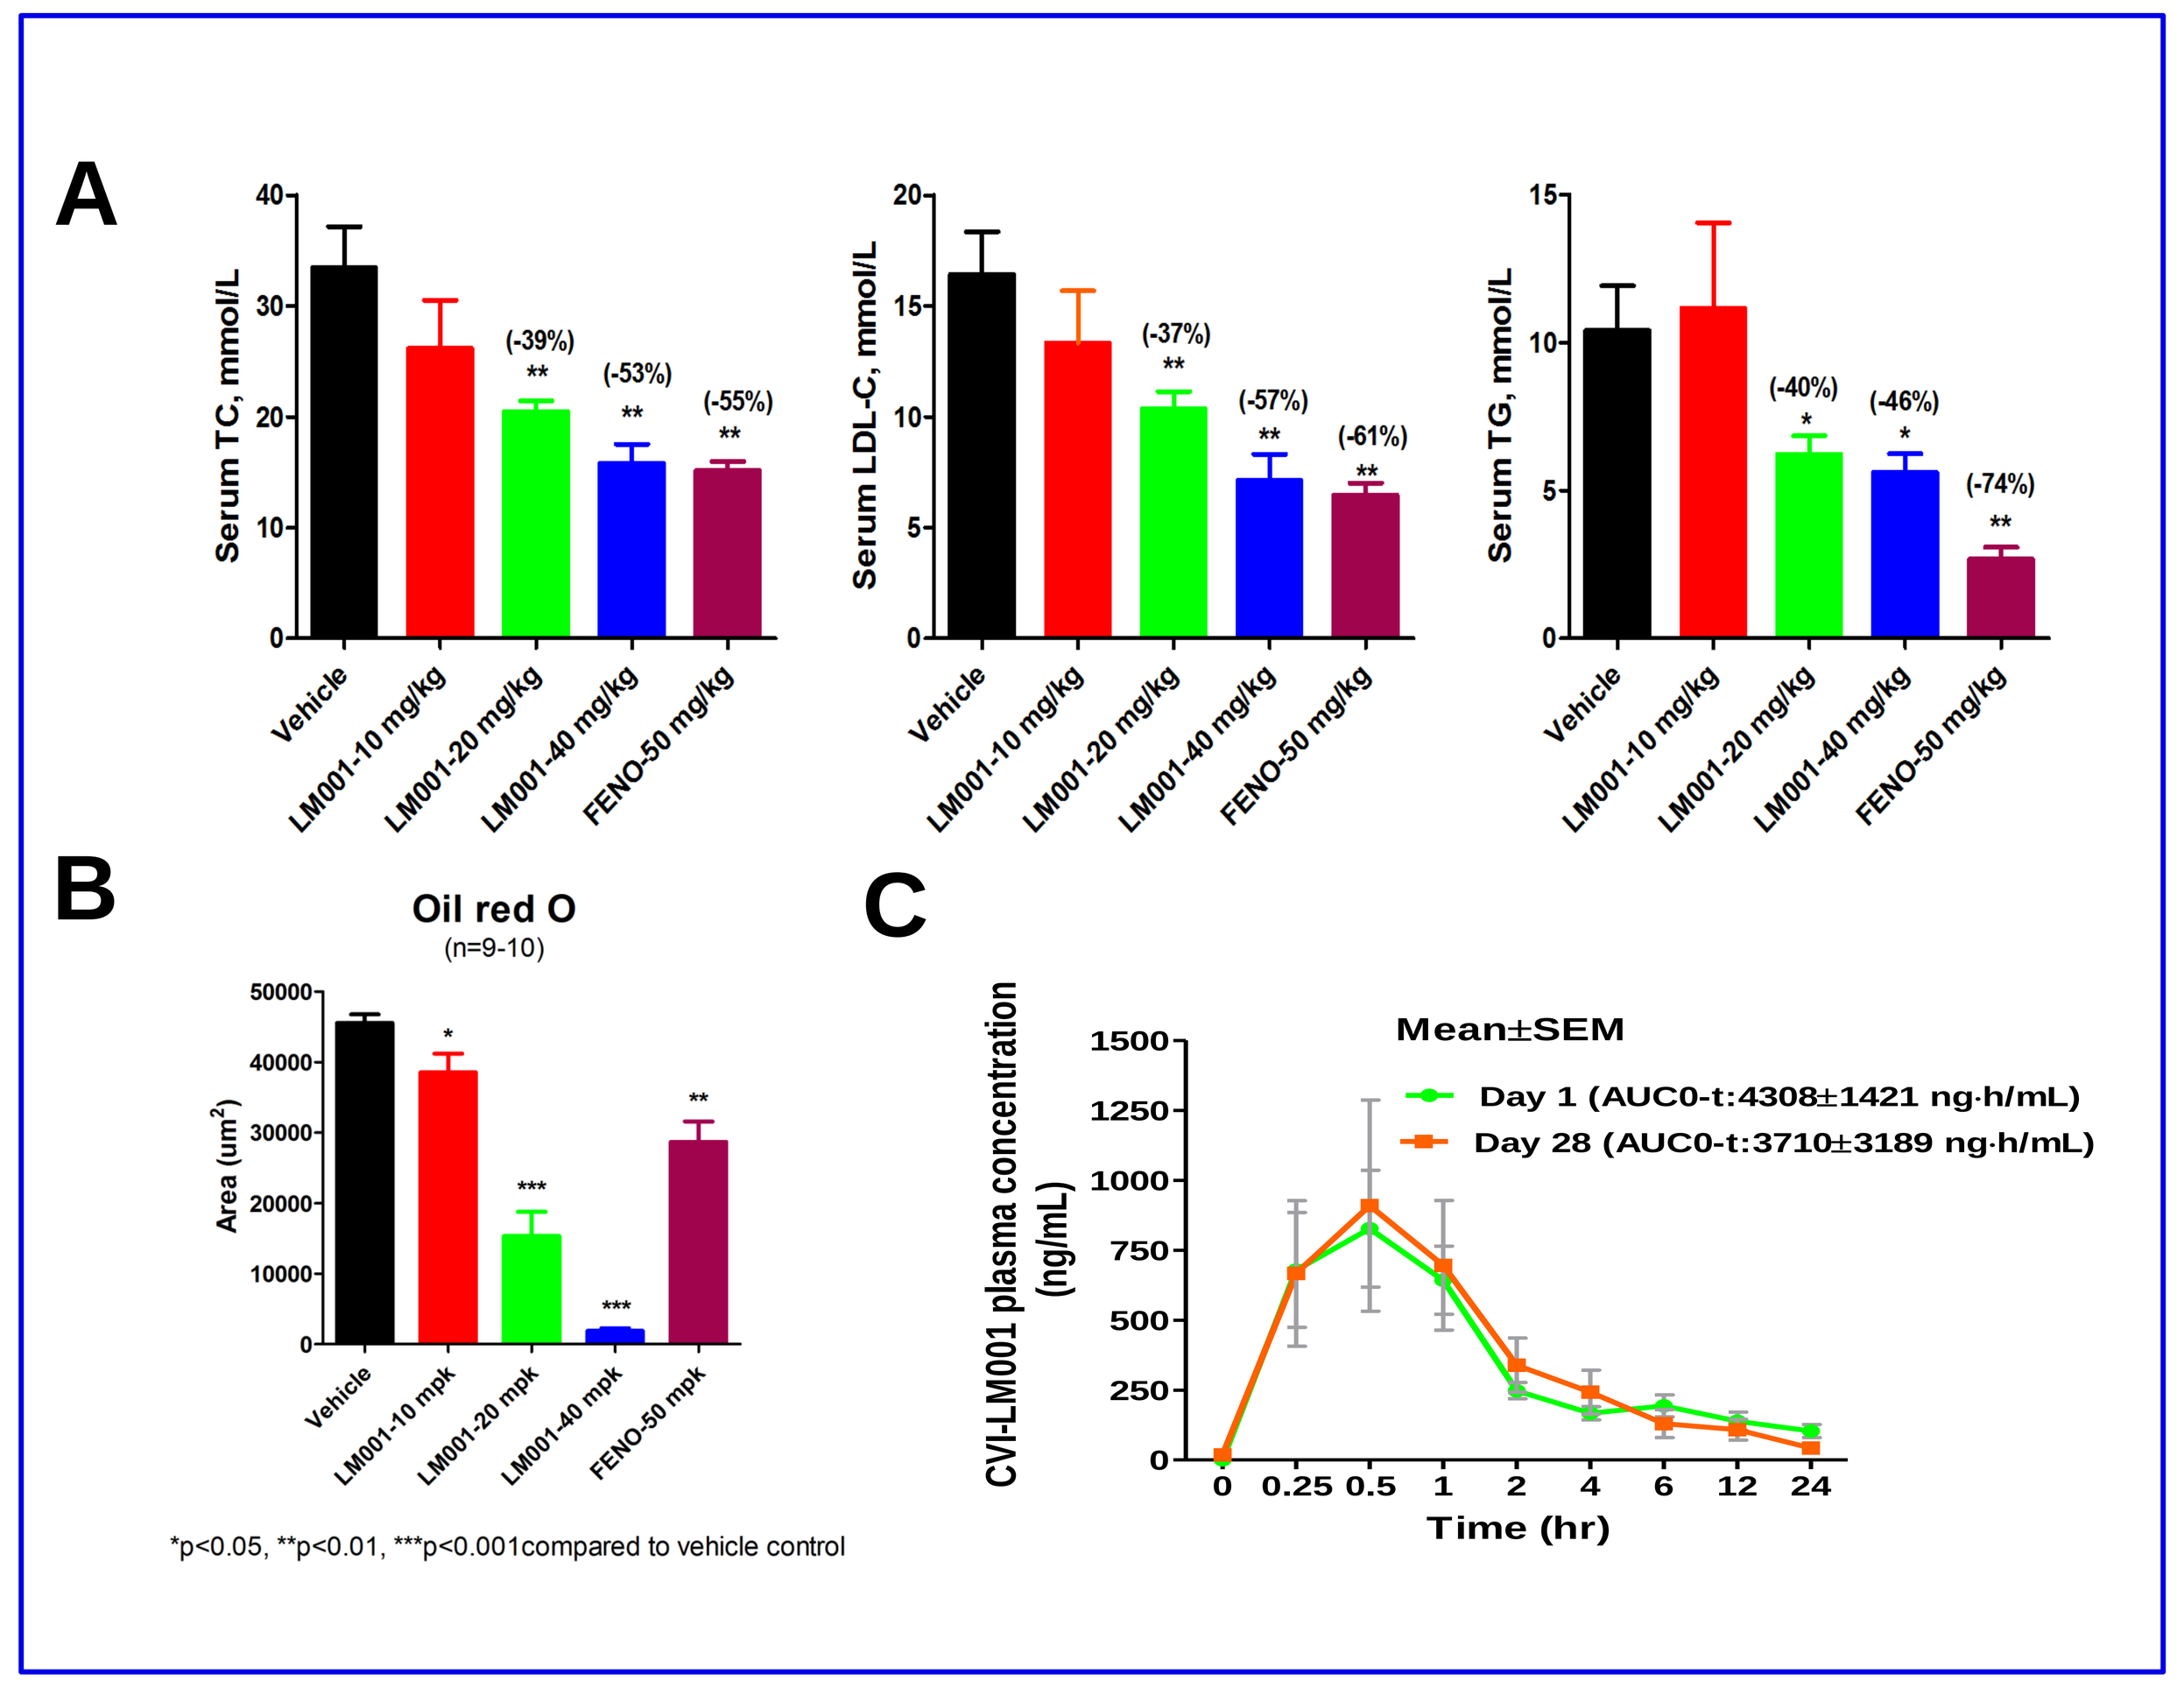
A
Serum TC, mmol/L
(-39%) (-53%) (-55%)
Serum LDL-C, mmol/L
(-37%) (-57%) (-61%)
Serum TG, mmol/L
(-40%) (-46%) (-74%)
Vehicle
LM001-10 mg/kg
LM001-20 mg/kg
LM001-40 mg/kg
FENO-50 mg/kg
B
Oil red O
(n=9-10)
Area (um²)
C
Mean±SEM
Day 1 (AUC0-t:4308±1421 ng·h/mL)
Day 28 (AUC0-t:3710±3189 ng·h/mL)
CVI-LM001 plasma concentration (ng/mL)
Time (hr)
*p<0.05, **p<0.01, ***p<0.001compared to vehicle control

Figure 2. In the free-choice diet-induced NASH hamster model, CVI-LM001 showed benefits on NASH, with significantly lower fibrosis and hepatocyte ballooning and a trend towards lower steatosis and inflammation score, resulting in a significant reduction in NAS score, an effect also observed with the reference drug elafibranor (ELA).

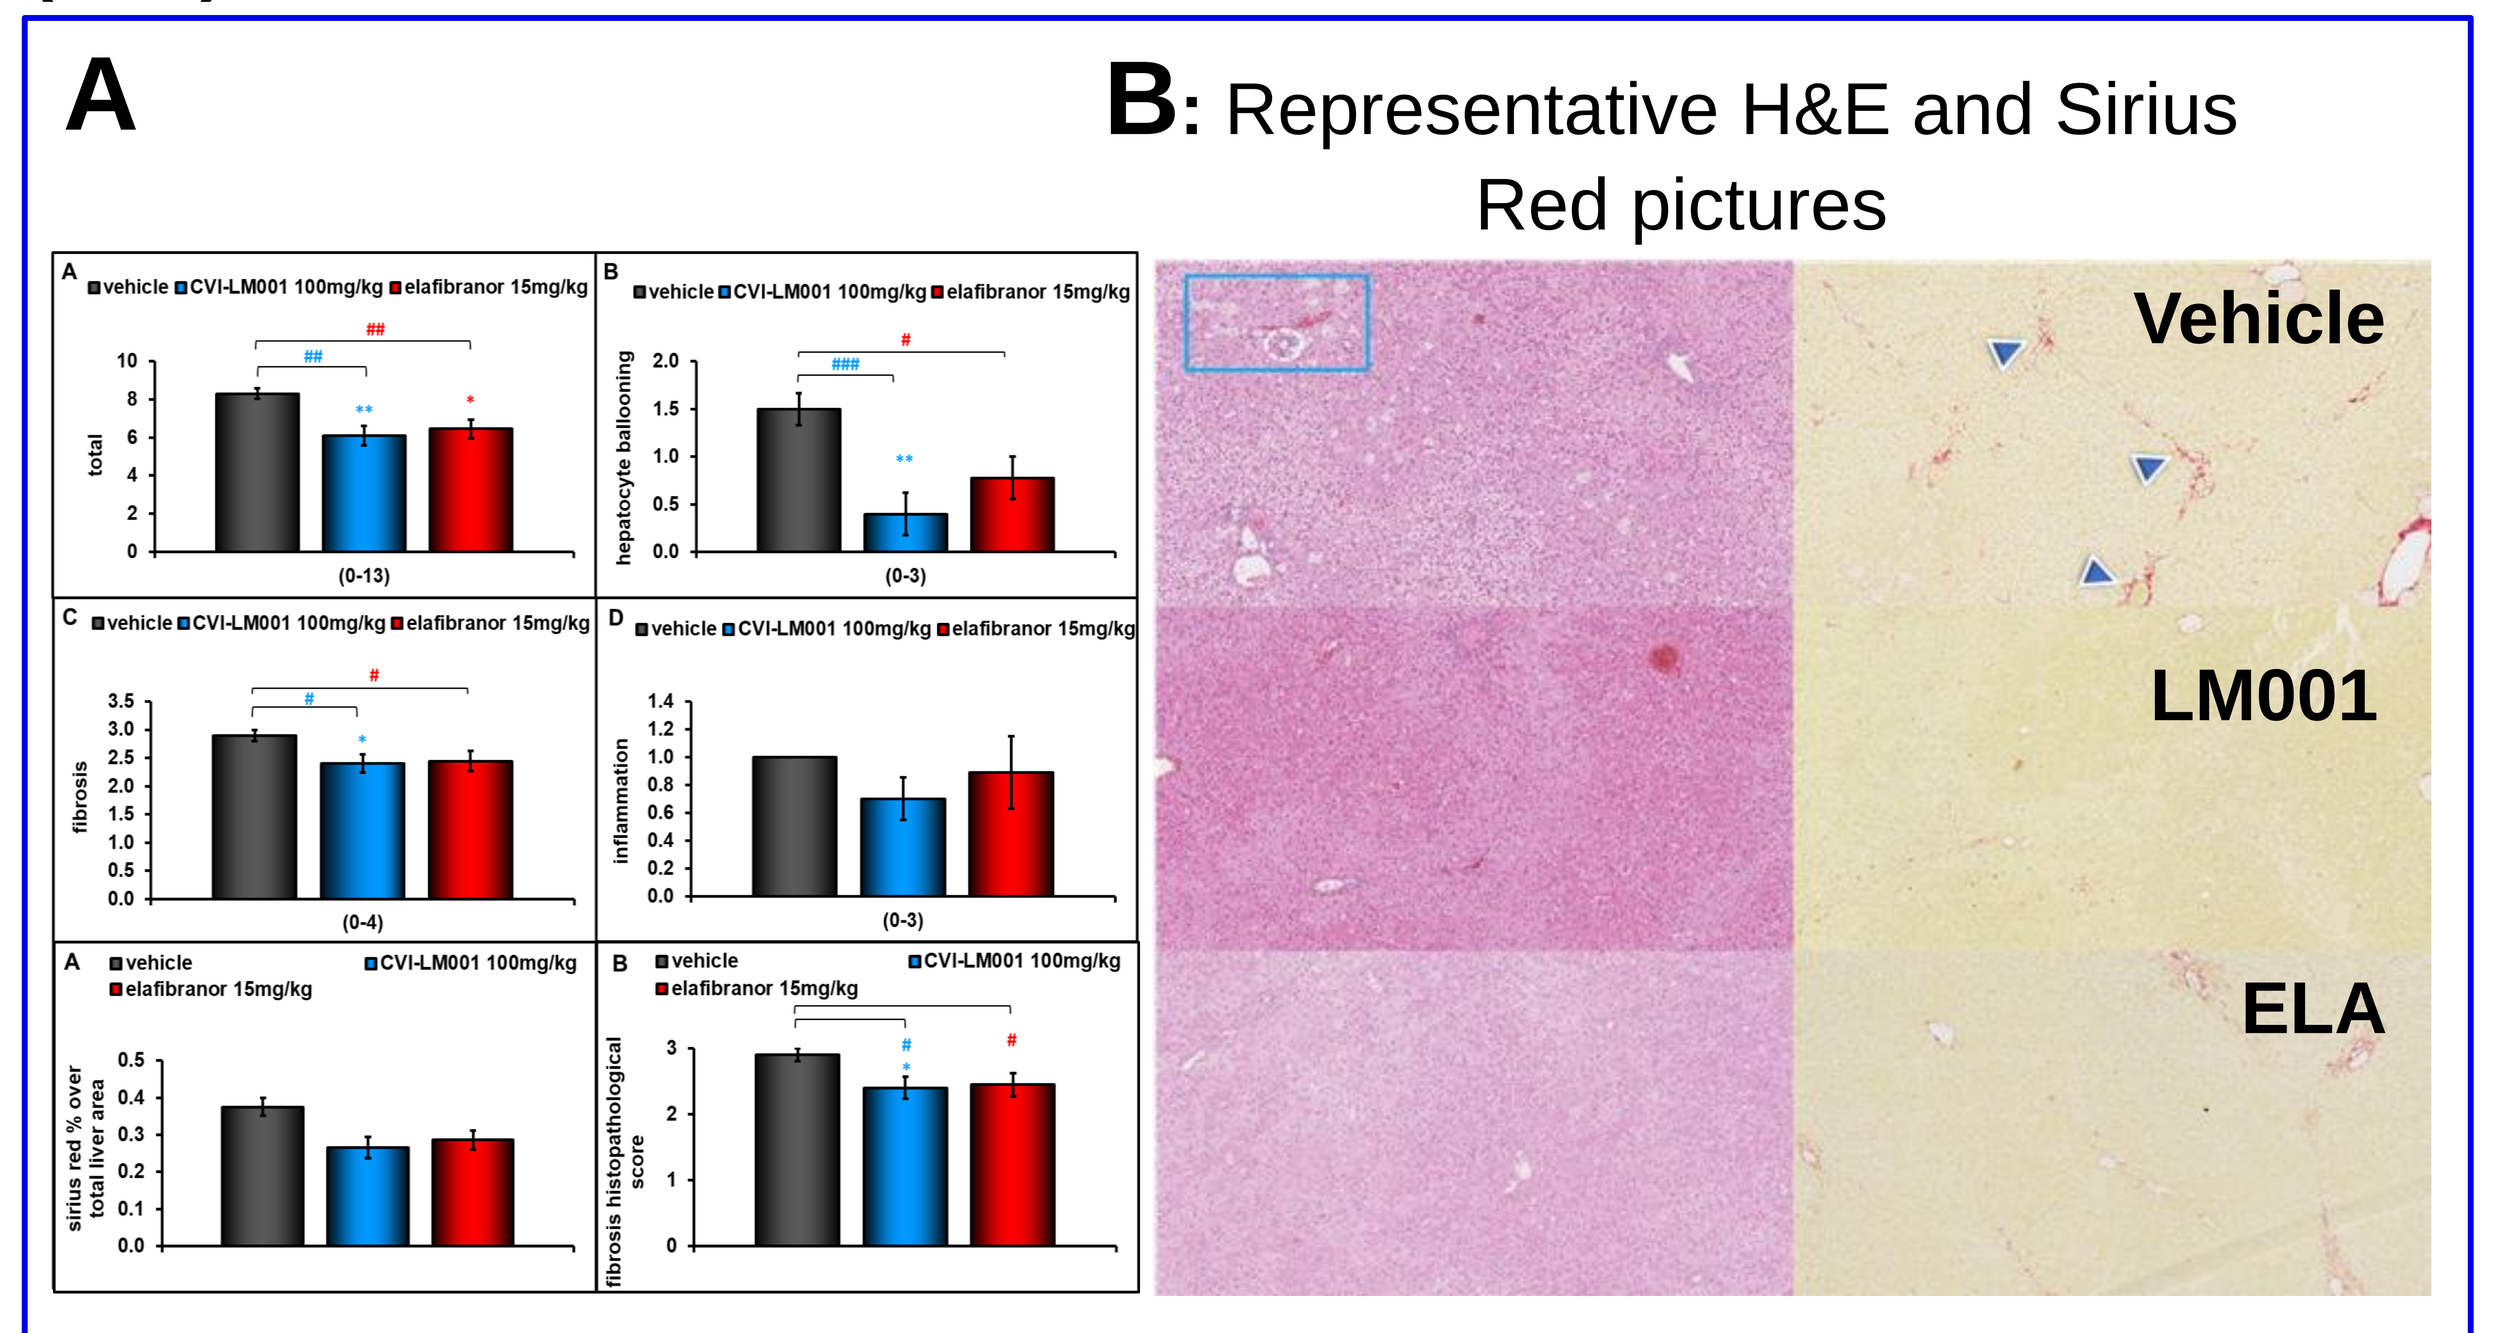
A
B: Representative H&E and Sirius Red pictures
vehicle CVI-LM001 100mg/kg elafibranor 15mg/kg
total
hepatocyte ballooning
fibrosis
inflammation
Vehicle
LM001
ELA
Vehicle
CVI-LM001
Elafibranor

C: Representative H&E pictures (x20). Blue squares indicate microvesicular steatosis with several ballooned hepatocytes with the presence of mixed cell inflammation (white circle).

Phase 1a Healthy subjects MAD Results

Figure 3. CVI-LM001 Phase Ia Clinical Highlights:

- Excellent safety profile of SAD (100-800 mg QD) and MAD (100-300 mg QD) studies and good plasma exposure (SAD: **A-D**; **E**: MAD) were observed;
- Ten day treatment with CVI-LM001 led to substantial reduction of blood PCSK9 levels **(F)**, indicating target engagement and validated the MOA of PCSK9 inhibition. The first-in-human PK data supports the once a day (QD) dosing regime in late stage of clinical development

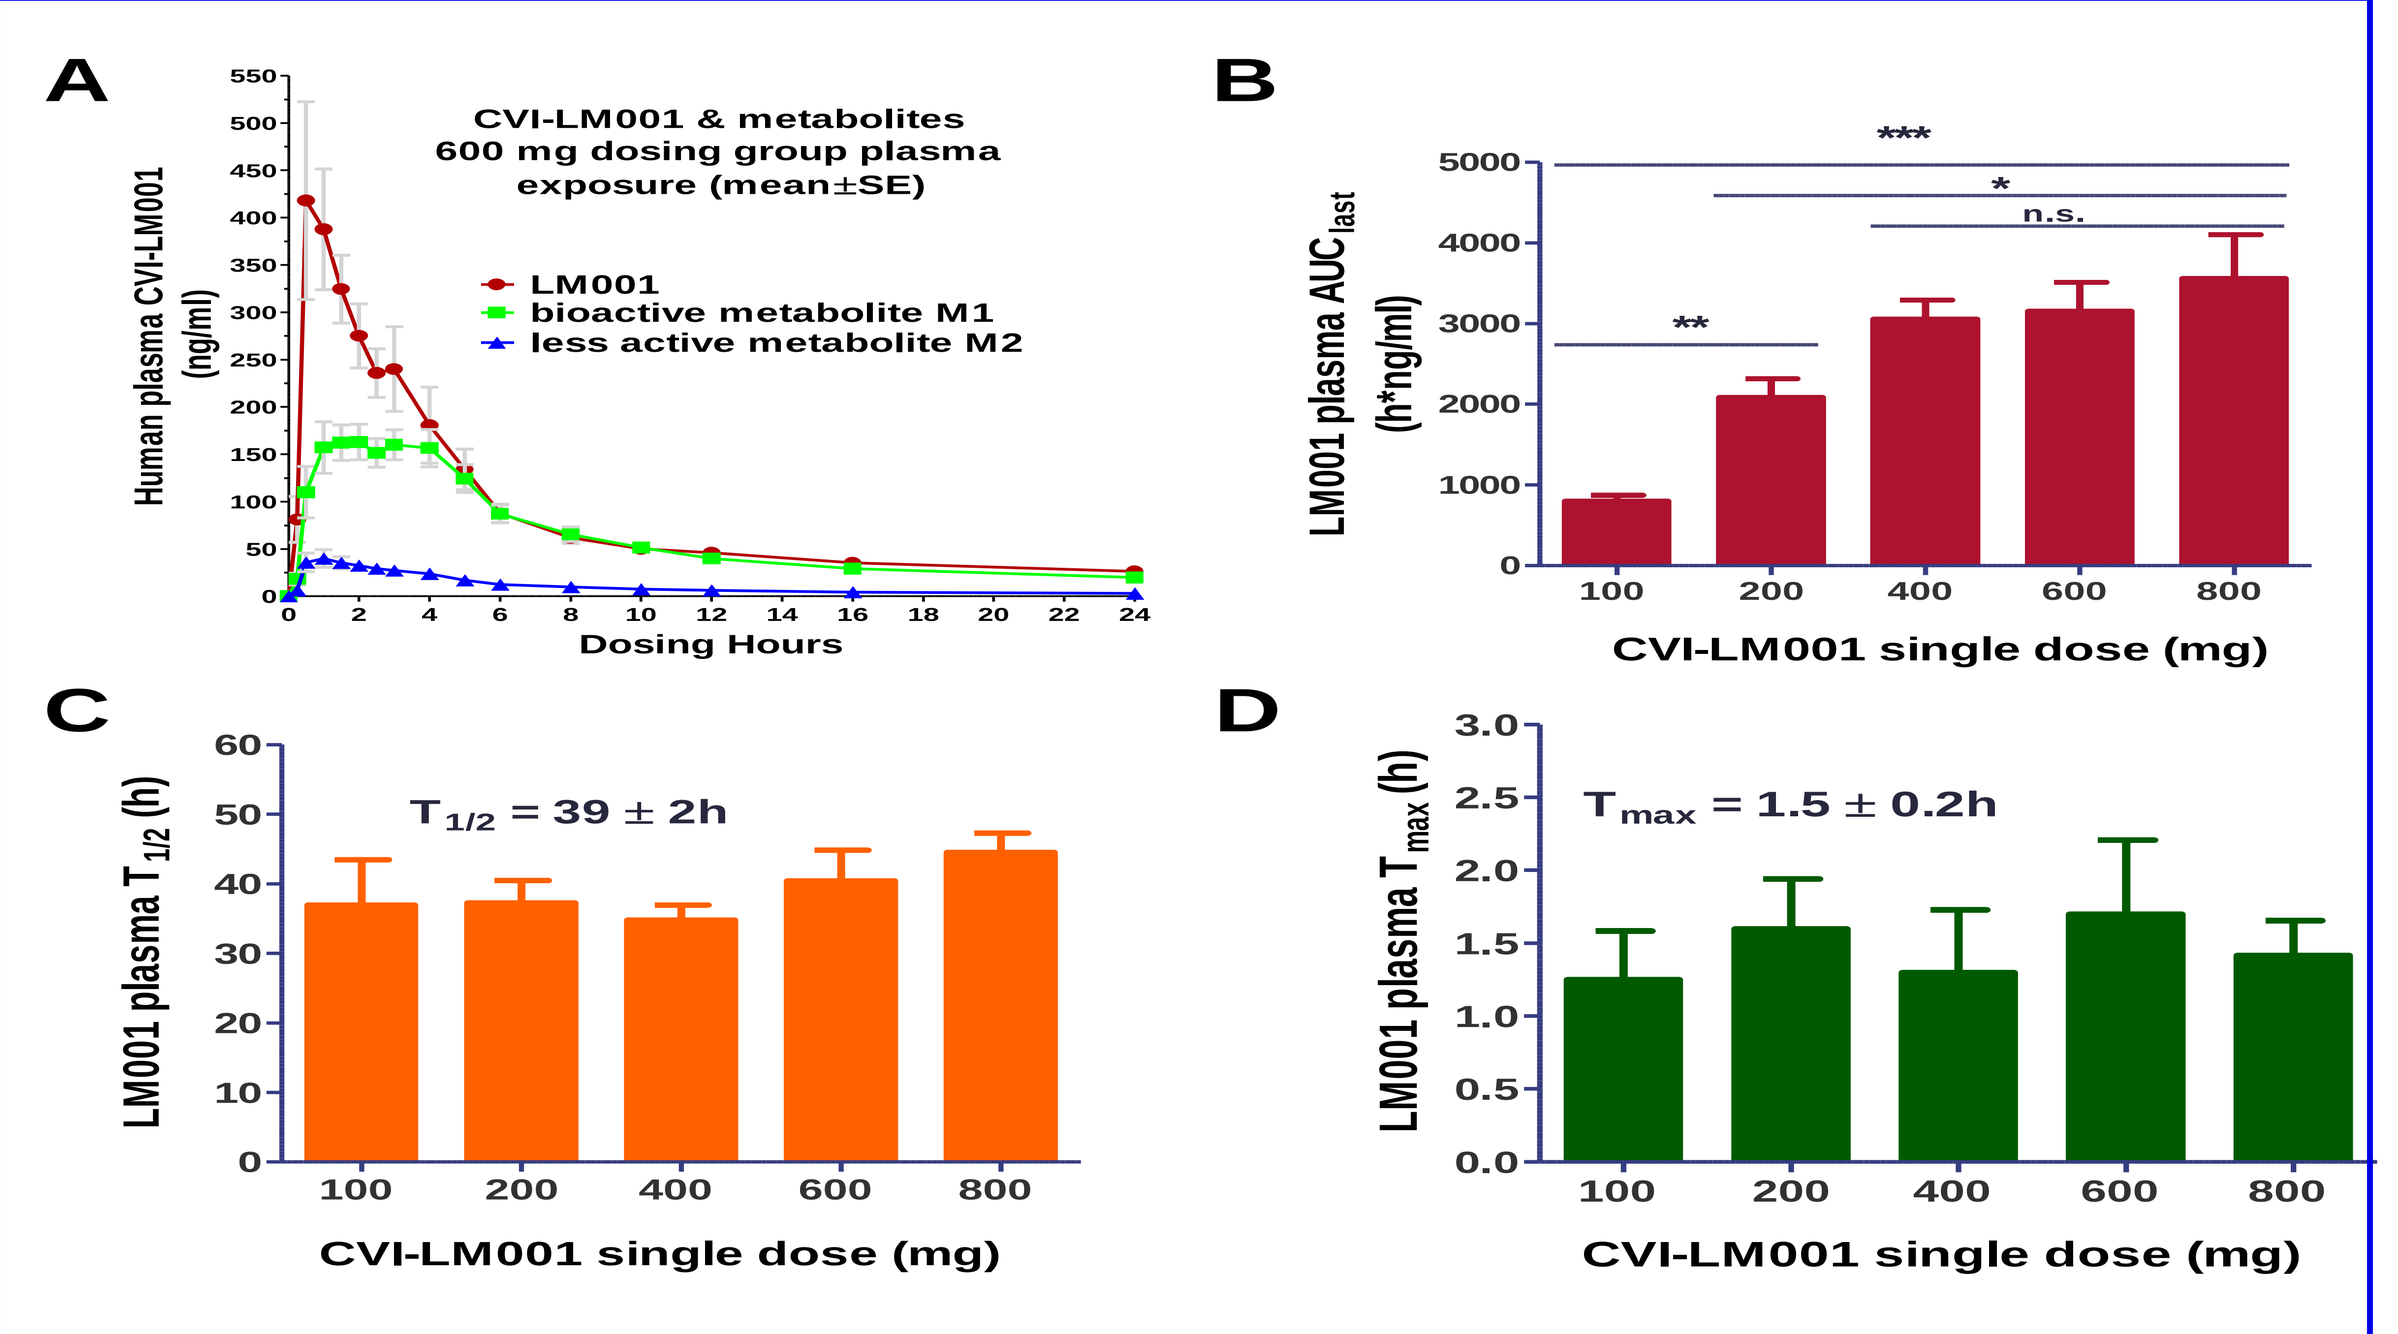
A
CVI-LM001 & metabolites
600 mg dosing group plasma exposure (mean±SE)
LM001
bioactive metabolite M1
less active metabolite M2
Human plasma CVI-LM001 (ng/ml)
Dosing Hours
B
LM001 plasma AUClast (h*ng/ml)
CVI-LM001 single dose (mg)
C
LM001 plasma T1/2 (h)
T1/2 = 39 ± 2h
CVI-LM001 single dose (mg)
D
LM001 plasma Tmax (h)
Tmax = 1.5 ± 0.2h
CVI-LM001 single dose (mg)

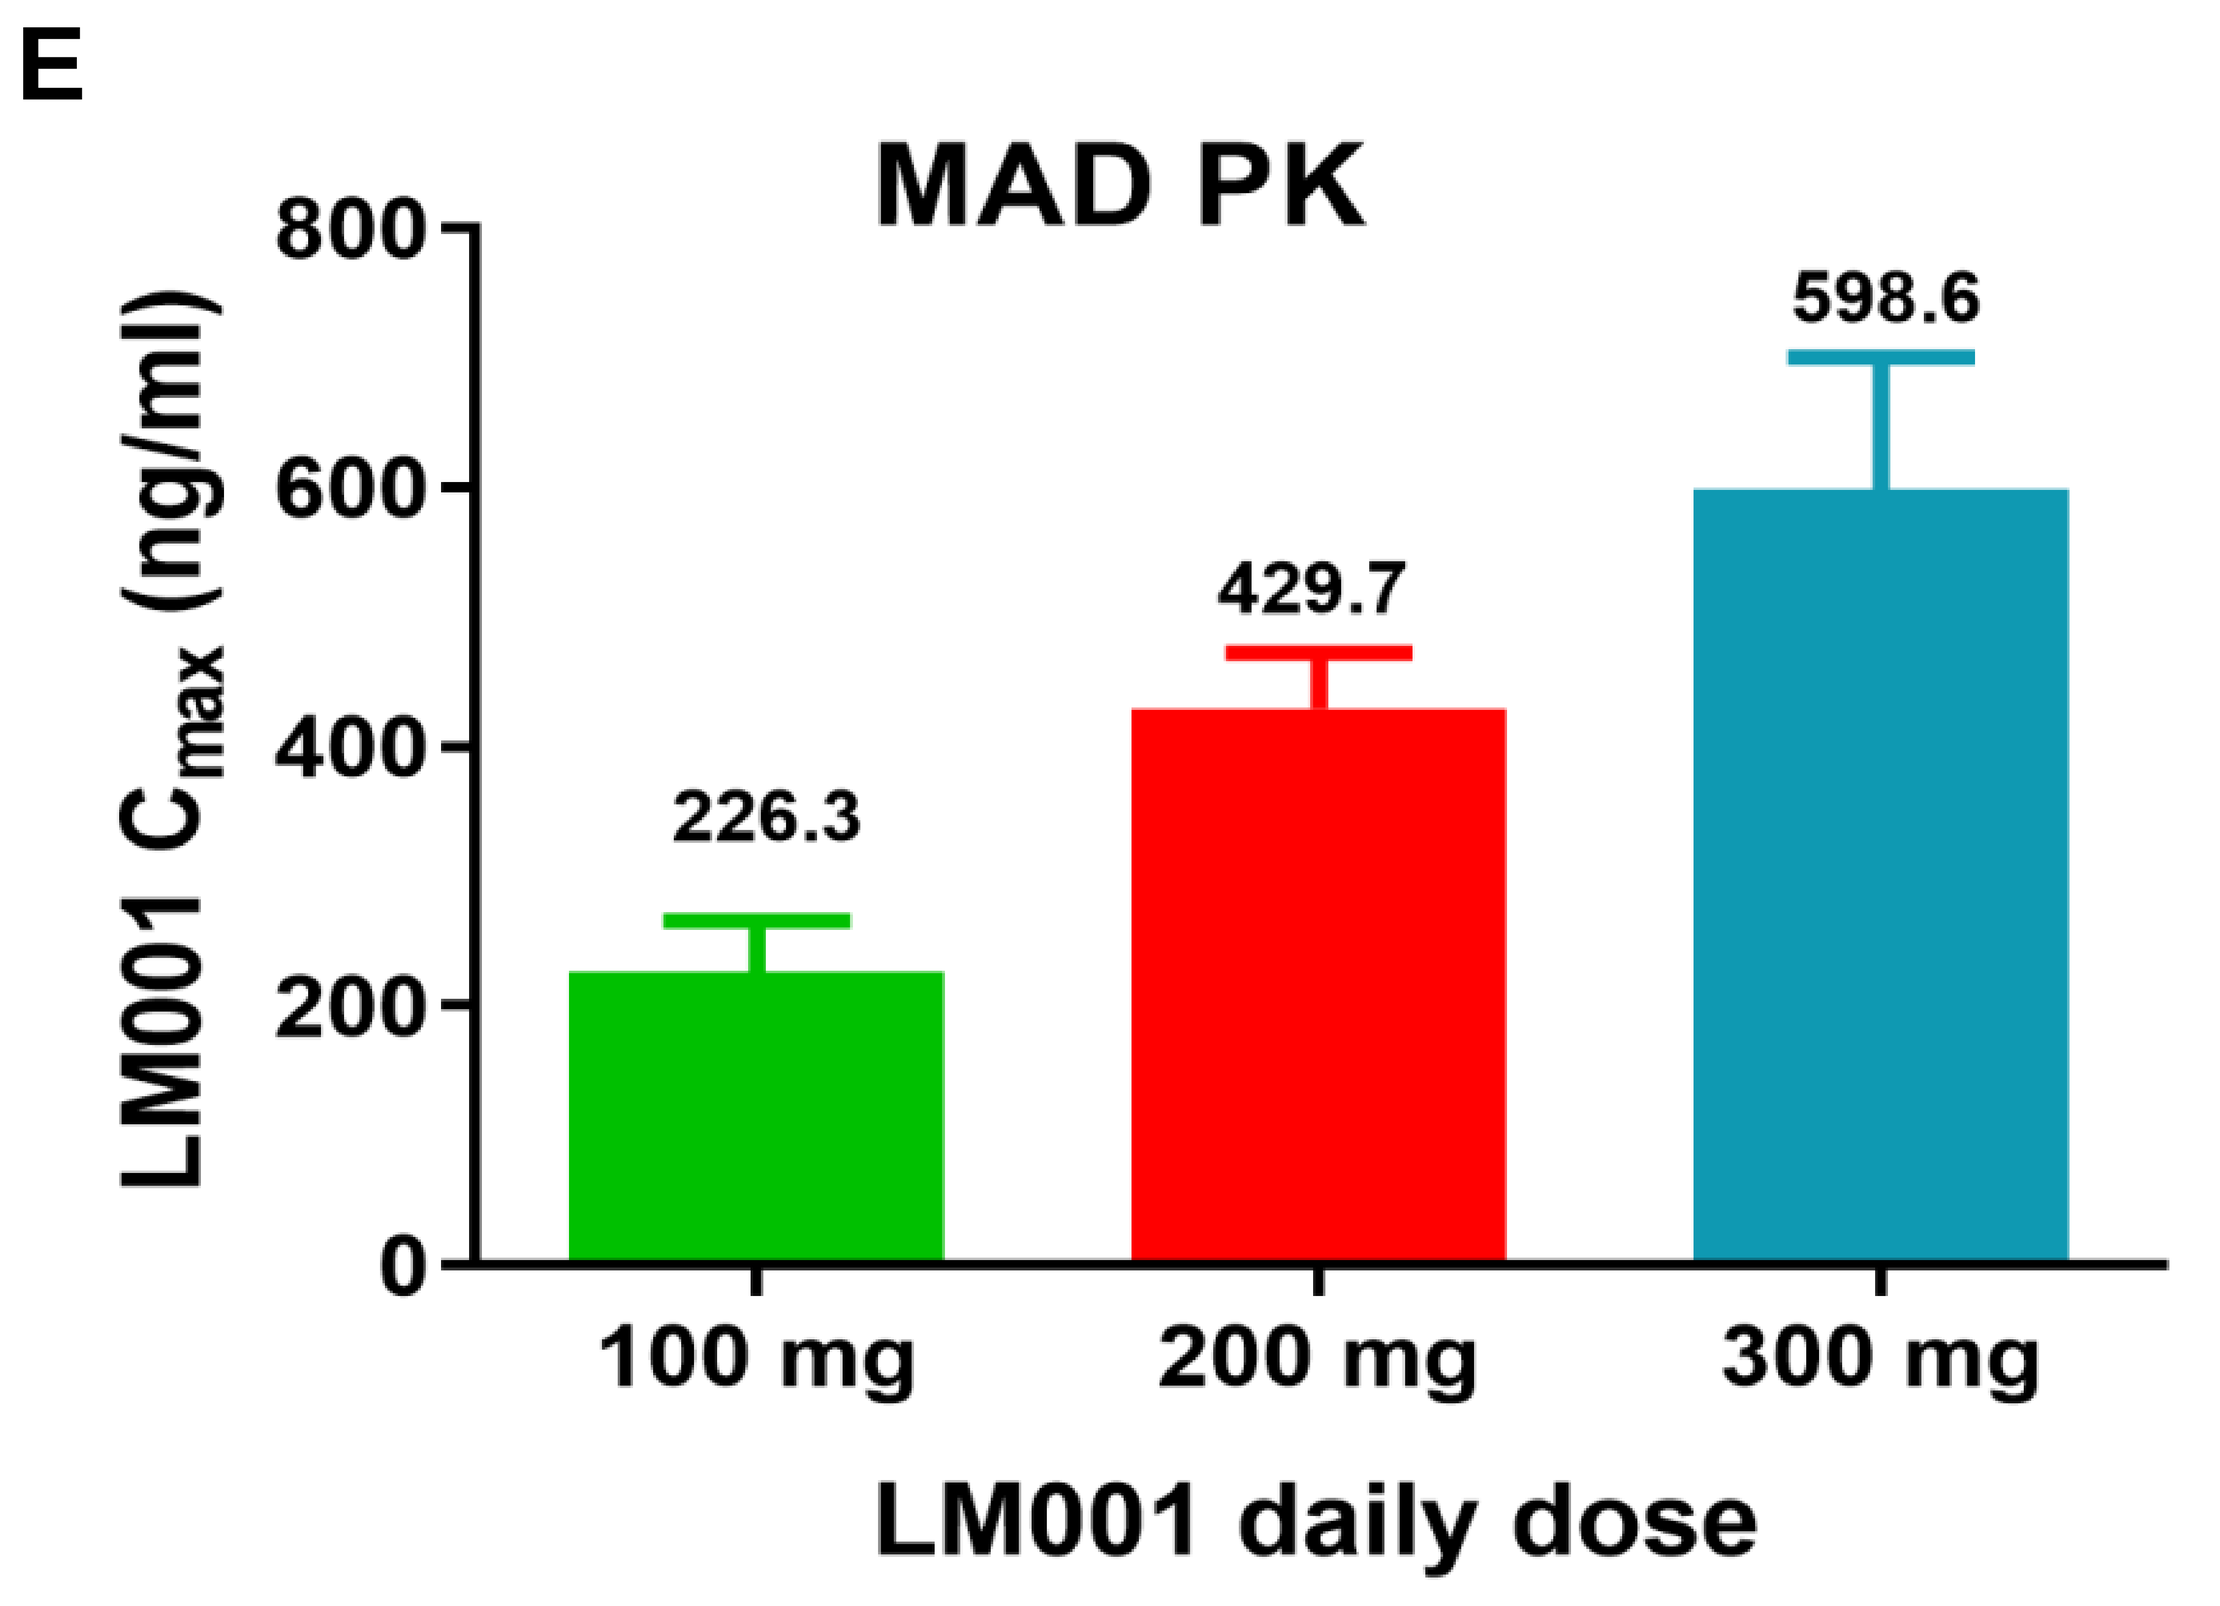
E
MAD PK
LM001 Cmax (ng/ml)
226.3
429.7
598.6
100 mg
200 mg
300 mg
LM001 daily dose

F: CVI-LM001 treatment of 10 days significantly reduced PCSK9 serum levels

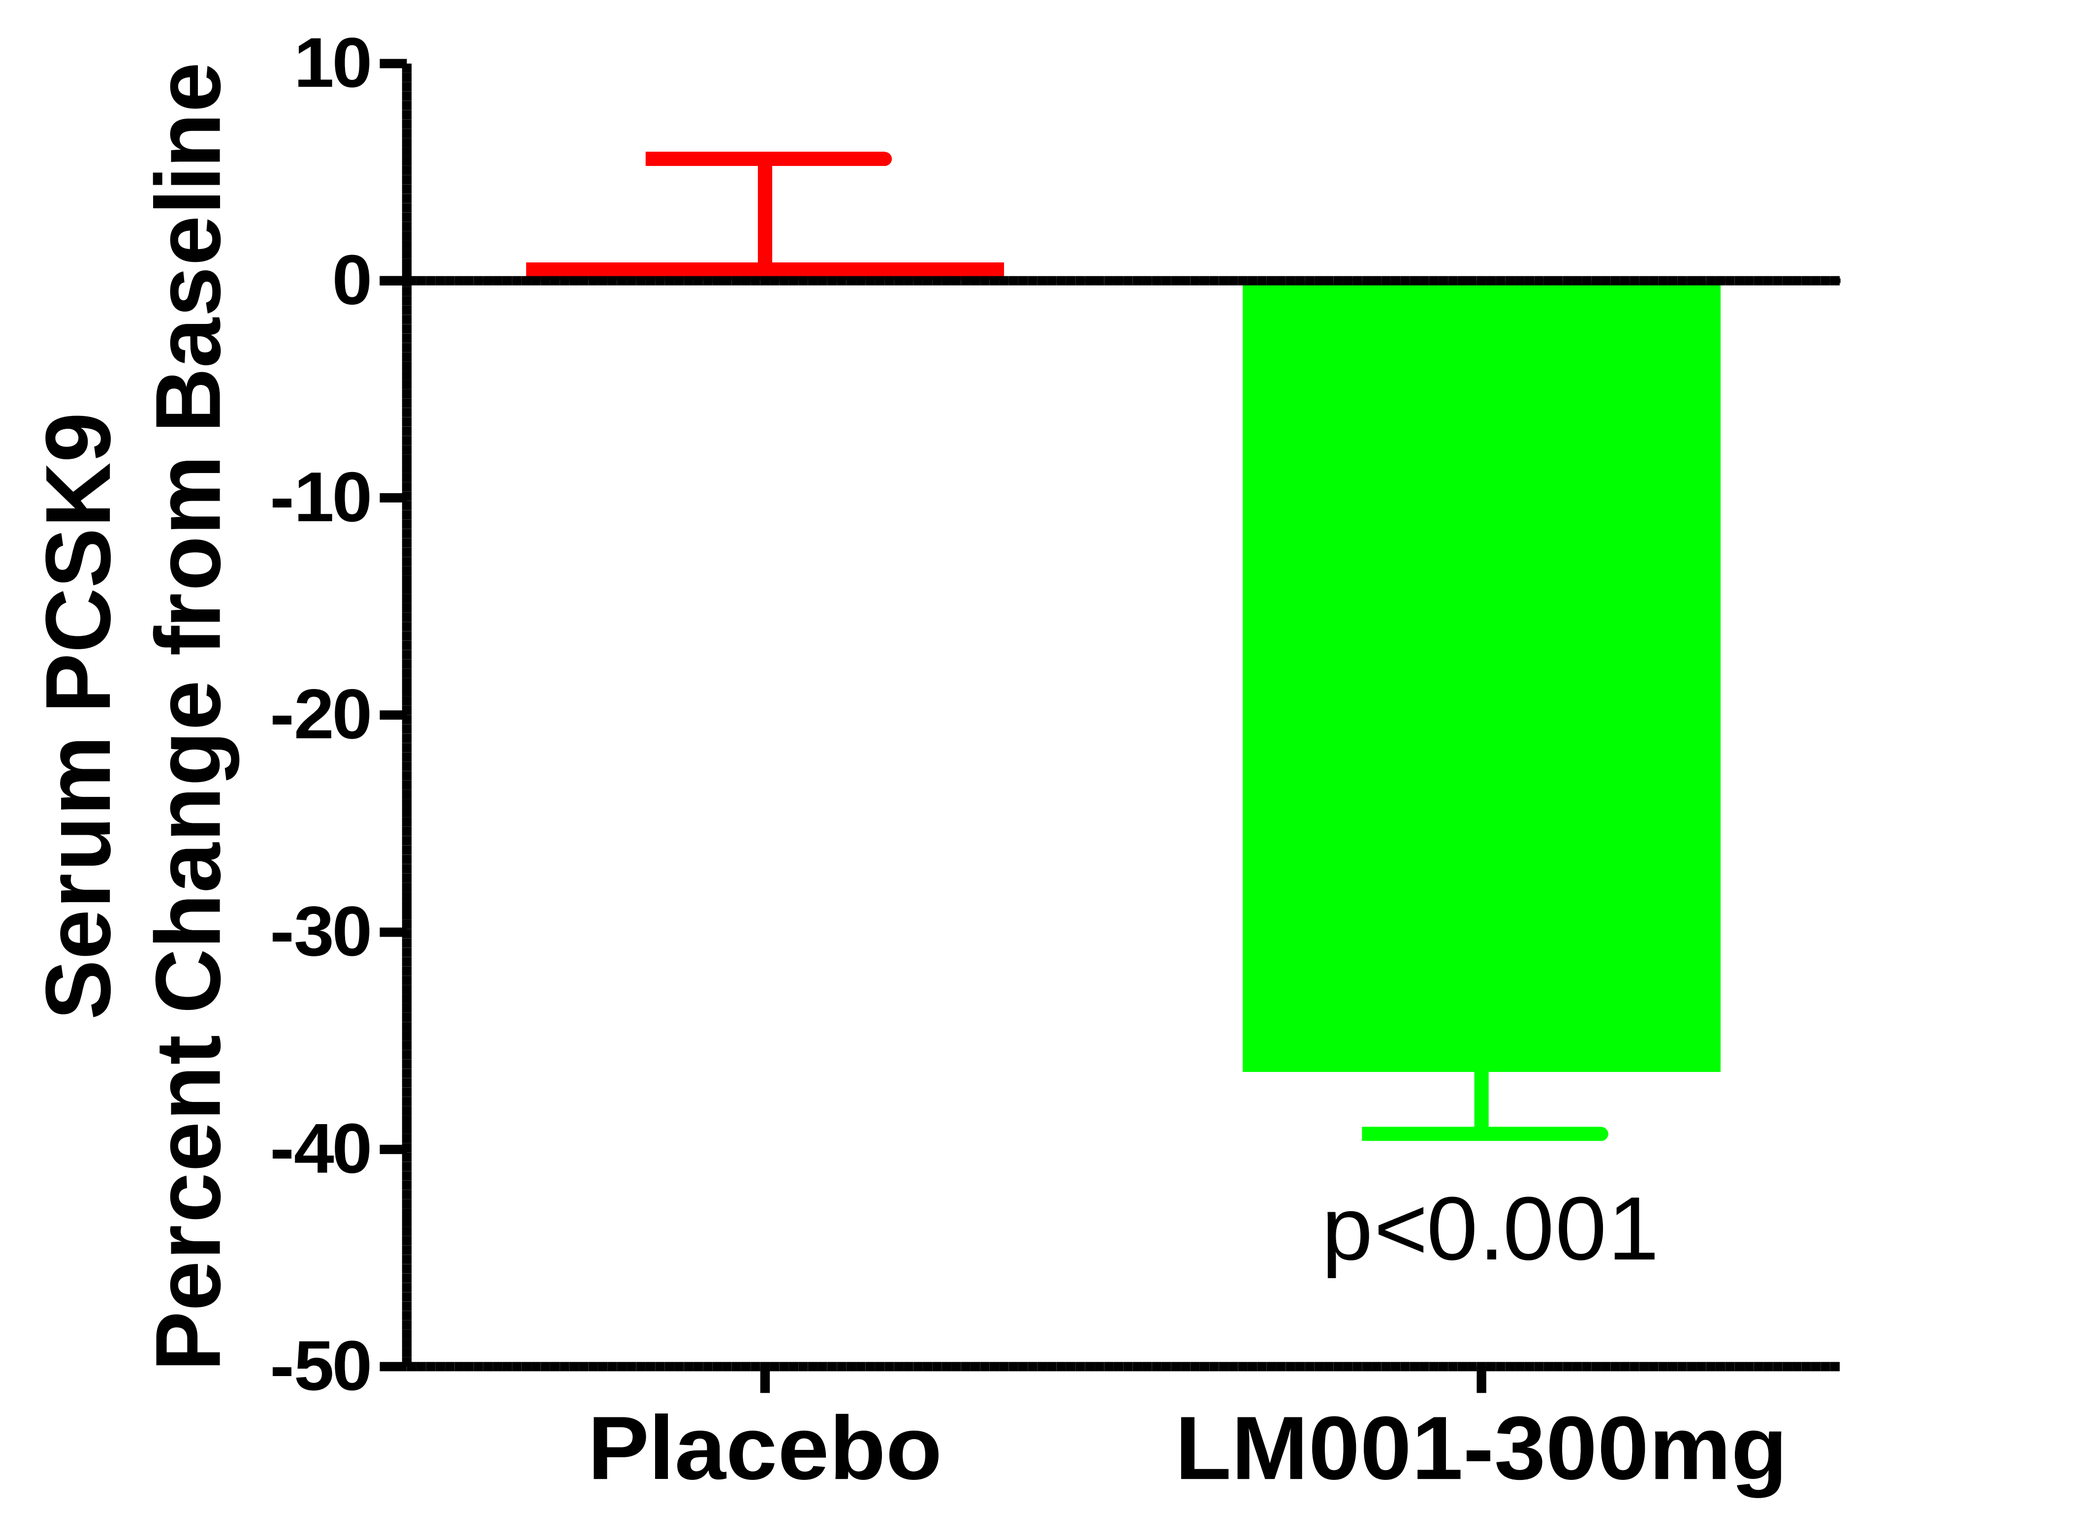
Serum PCSK9
Percent Change from Baseline
p<0.001
Placebo
LM001-300mg

Phase 1b Hyperlipidemic Subjects Results

Figure 4. CVI-LM001 28-Day Phase Ib summary: Randomized, double-blind and placebo-controlled trial design **(A)**; benign safety and tolerability profile, no SAEs; Statistically significant reductions in LDL-C, TC and ApoB by 300 mg daily dose **(B)**; CVI-LM001 200 mg dose showed trend in LDL-C and TC reductions; CVI-LM001 300 mg dose reduced PCSK9 serum levels in hyperlipidemic subjects which were consistent with LDL-C lowering **(C)**; Results support a Phase 2 12 week proof-of-concept (POC) study in hyperlipidemic subjects, which is currently undergoing.

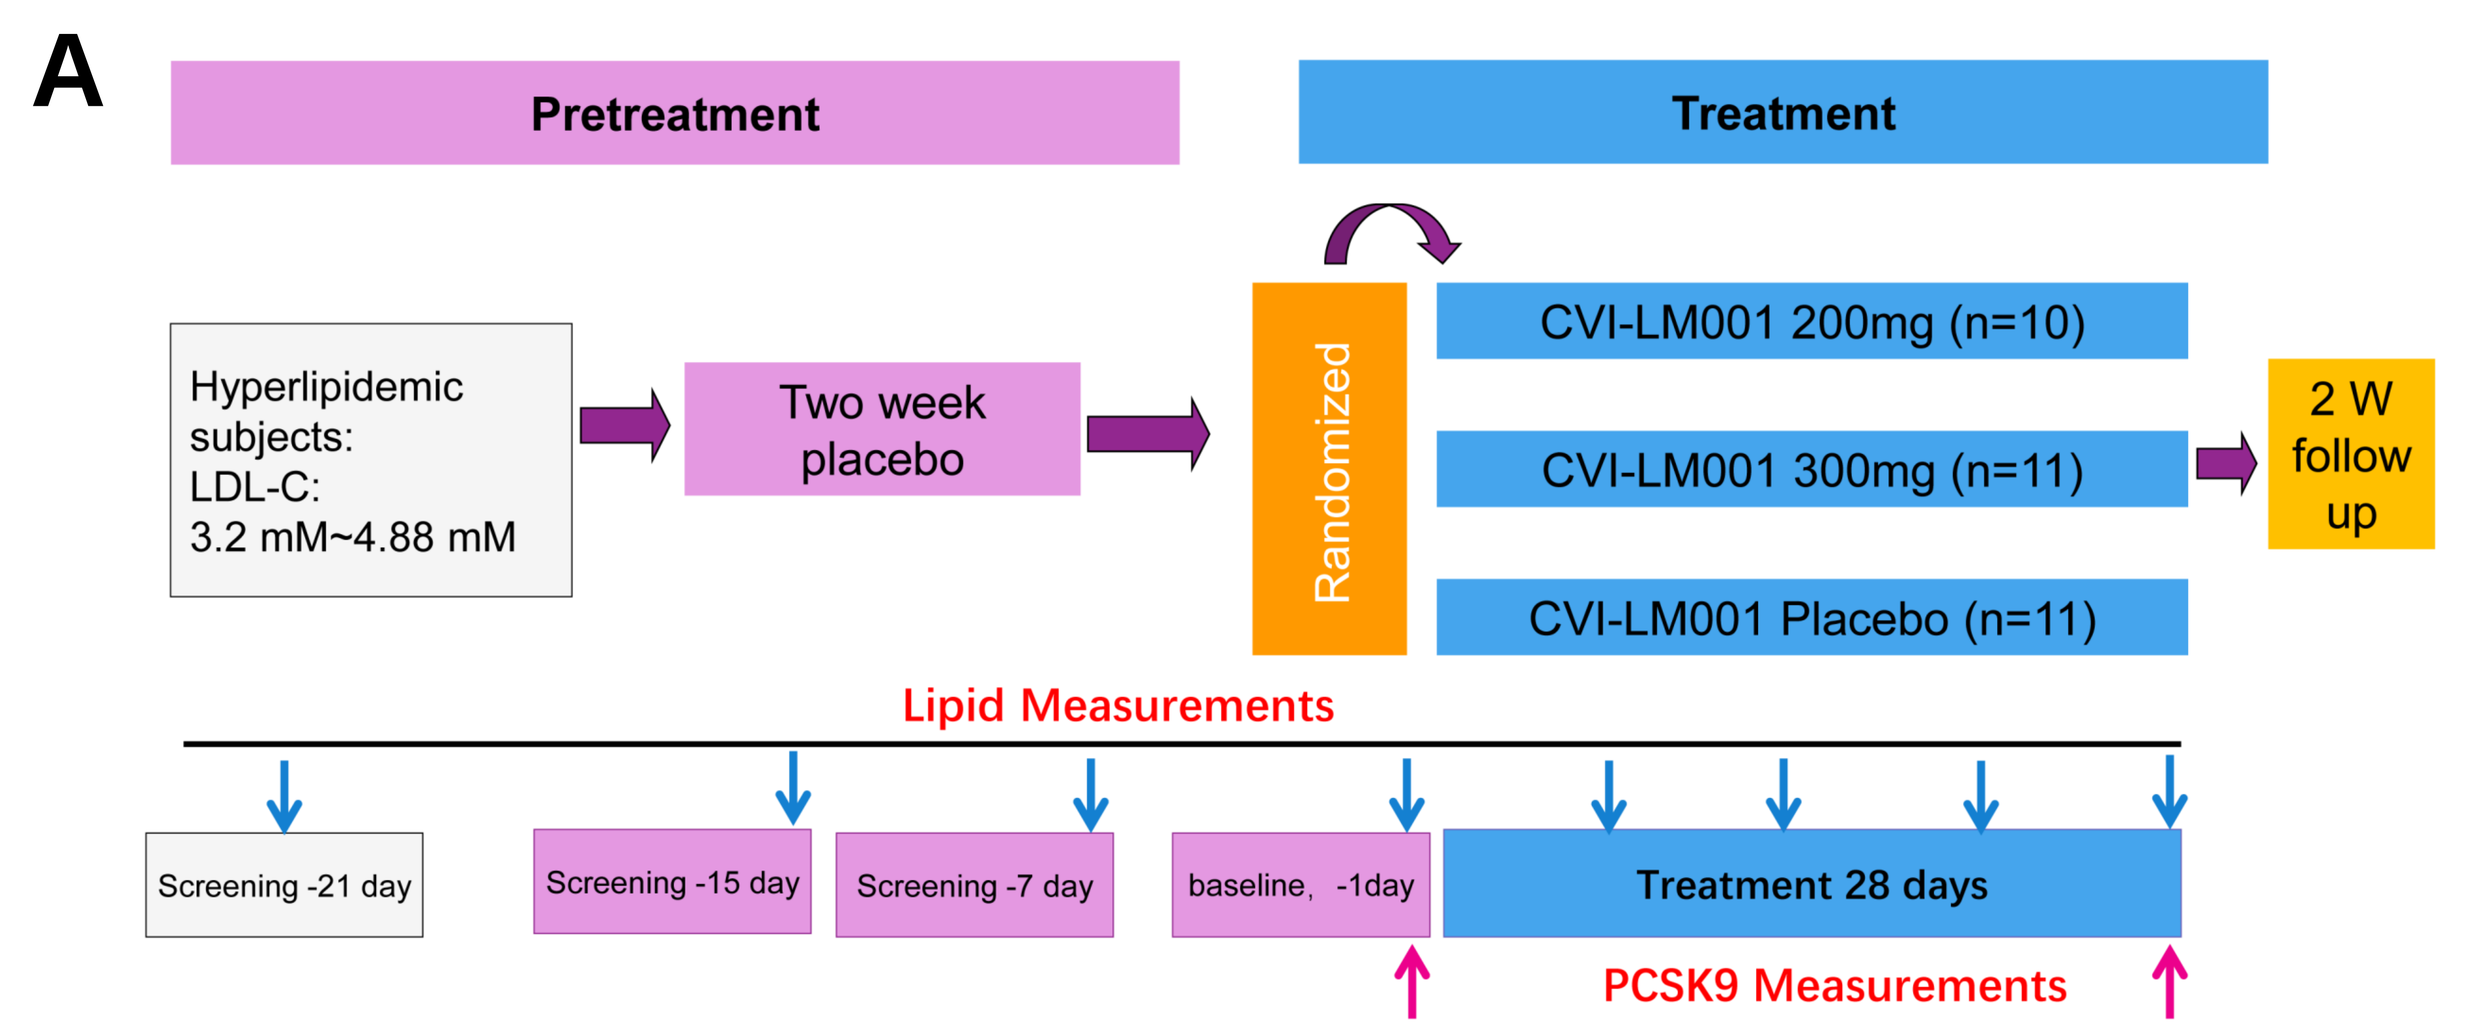
A
Pretreatment
Treatment
Hyperlipidemic subjects:
LDL-C:
3.2 mM~4.88 mM
Two week placebo
Randomized
CVI-LM001 200mg (n=10)
CVI-LM001 300mg (n=11)
CVI-LM001 Placebo (n=11)
2 W follow up
Lipid Measurements
Screening -21 day
Screening -15 day
Screening -7 day
baseline, -1day
Treatment 28 days
PCSK9 Measurements

B Placebo-adjusted changes from baseline (%)

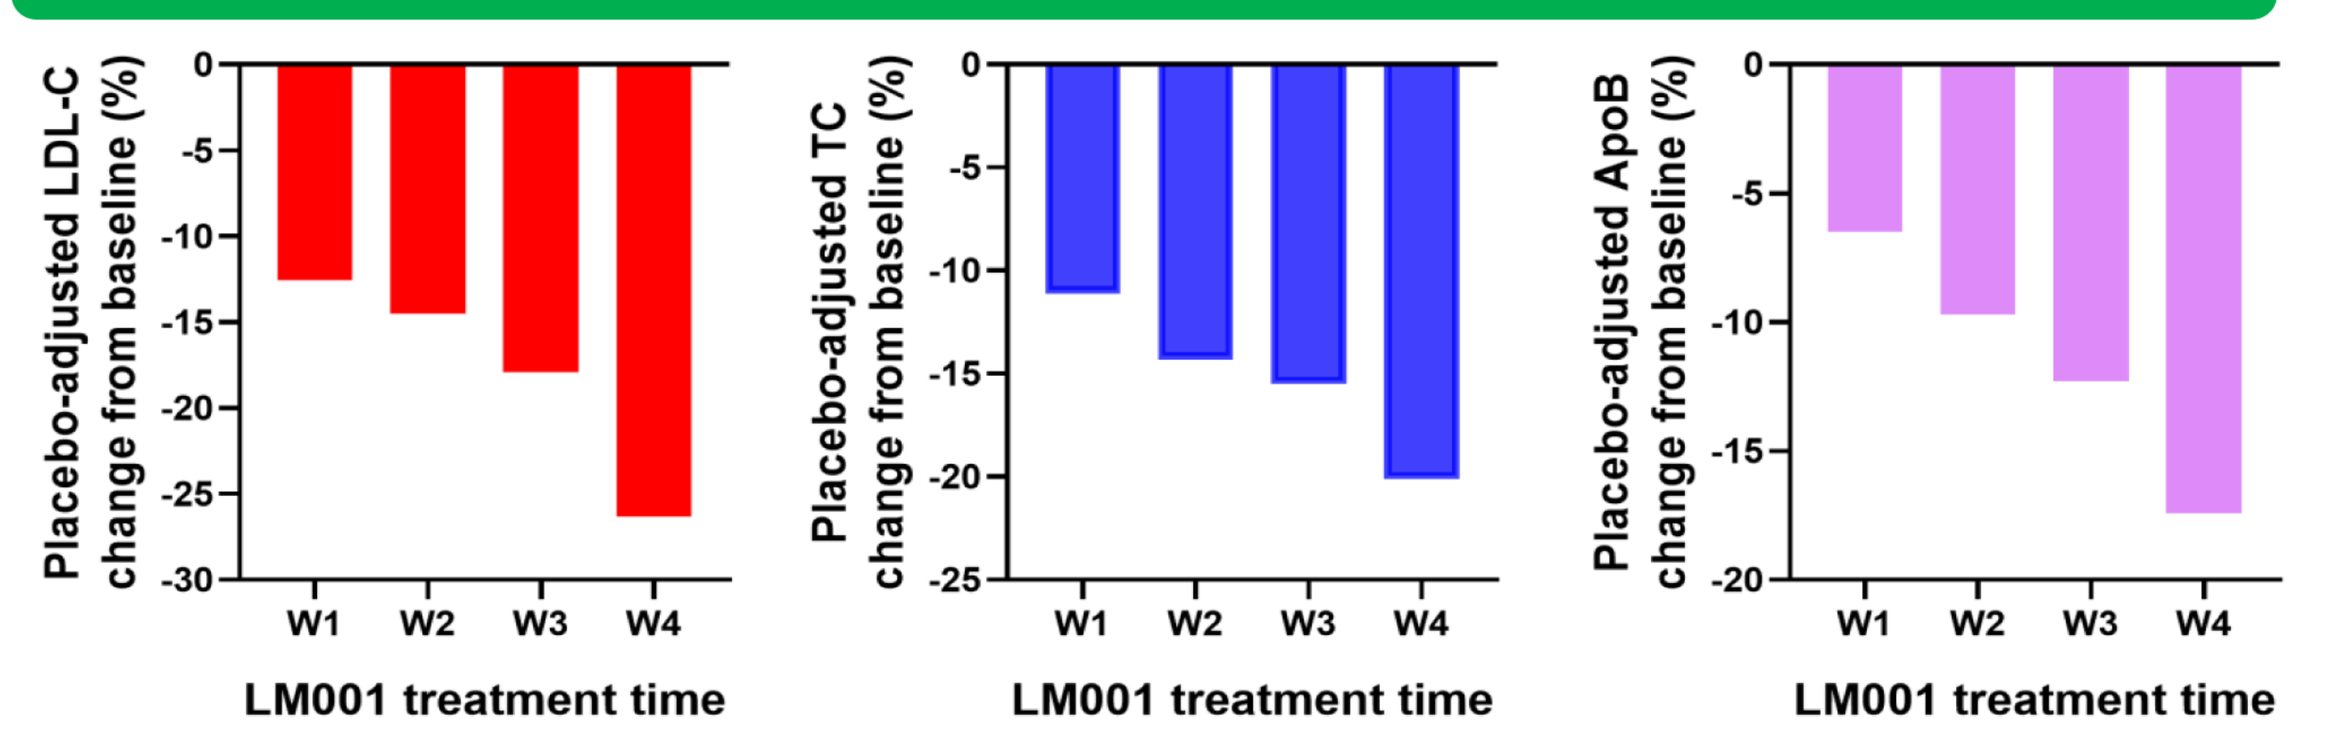
Placebo-adjusted LDL-C change from baseline (%)
Placebo-adjusted TC change from baseline (%)
Placebo-adjusted ApoB change from baseline (%)
W1
W2
W3
W4
LM001 treatment time

CVI-LM001 (300 mg, QD)	W1	W2	W3	W4
Placebo adjusted reduction, LDL-C:	-12.60%	-14.50%	-17.90%	-26.30%
	p=0.03	p=0.03	p=0.1	p=0.006
Placebo adjusted reduction, TC:	-11.10%	-14.30%	-15.50%	-20.10%
	p=0.03	p=0.03	p=0.01	p=0.002
Placebo adjusted reduction, ApoB:	-6.50%	-9.70%	-12.30%	-17.40%
	p>0.5	p>0.05	p0.02	p=0.01

* One Subject in 300 mg group was found having rheumatoid arthritis (RA) before dosing and during the study, that RA subject data was not included

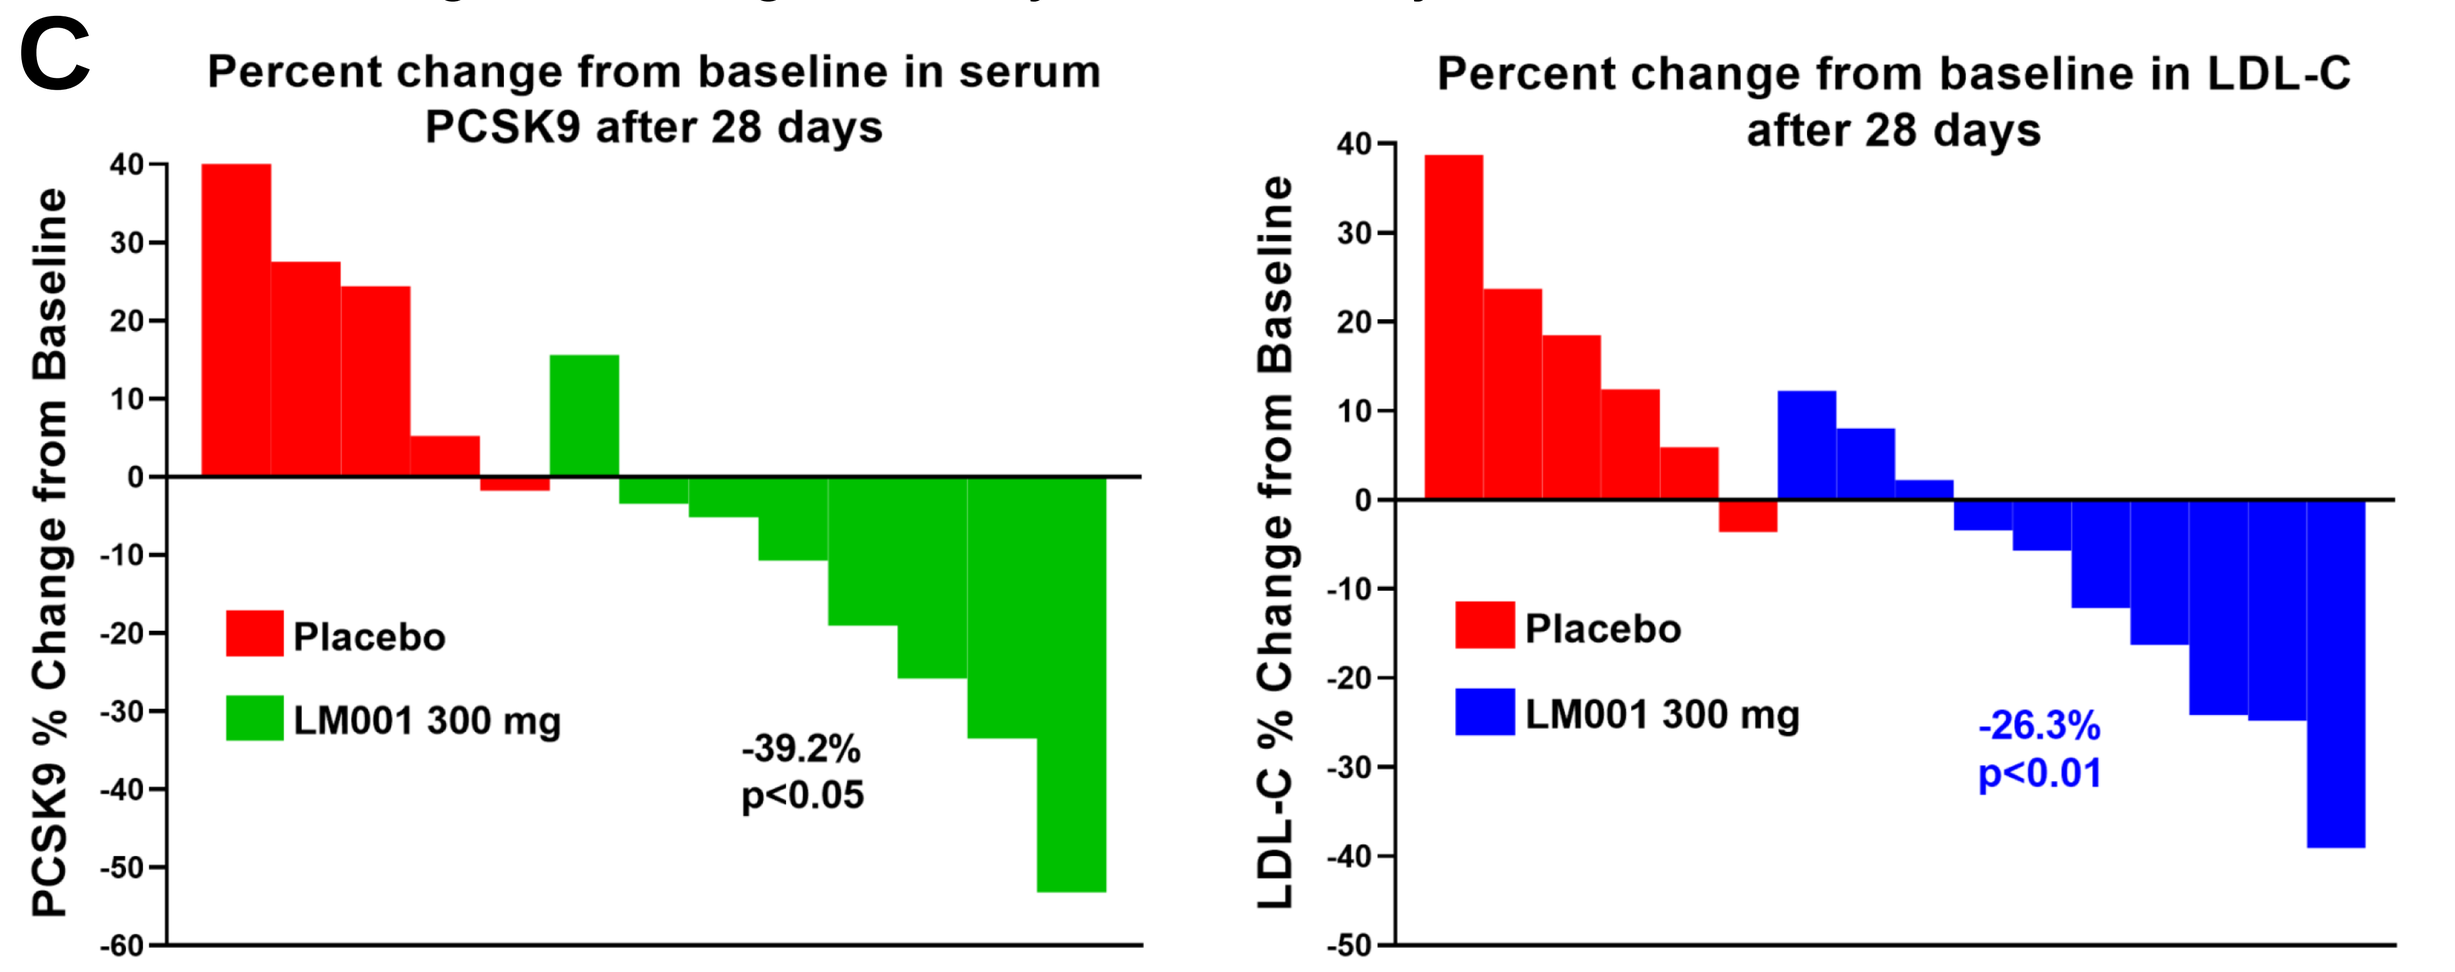
C
Percent change from baseline in serum PCSK9 after 28 days
PCSK9 % Change from Baseline
Placebo
LM001 300 mg
-39.2%
p<0.05
Percent change from baseline in LDL-C after 28 days
LDL-C % Change from Baseline
Placebo
LM001 300 mg
-26.3%
p<0.01

** Two subjects from LM001 cohort and one from placebo cohort had abnormal PCSK9-LDLC relationship and were excluded from this analysis

CONCLUSION

These studies demonstrate that CVI-LM001, a novel PCSK9 modulator, has the potential to be a new oral cholesterol-lowering drug to treat hypercholesterolemia and NAFLD/NASH.